FPT155-001: A Phase 1a/1b Study of FPT155 (CD80-Fc) in Patients with Advanced Solid Tumors

Poster # TPS42

Michael Millward[1], Hui Gan[2], Anthony Joshua[3], James Kuo[4], Amy Prawira[3], Gary Richardson[5], Susannah D. Barbee[6], Sandeep Inamdar[6], Kristen L. Pierce[6], Mona Qureshi[6], Lisa Horvath[7];

[1]Linear Clinical Research, Perth, Australia; [2]Olivia Newton John Cancer Research Institute, Victoria, Australia; [3]St. Vincent's Hospital, Sydney, Australia; [4]Scientia Clinical Research, Sydney, Australia; [5]Cabrini Health Australia, Melbourne, Australia; [6]Five Prime Therapeutics Inc., South San Francisco, CA, USA; [7]Chris O'Brien Lifehouse, Sydney, Australia

Introduction

- Despite recent advances in immunotherapy, including checkpoint inhibitors such as PD-(L)1 and CTLA-4 antibodies, only a fraction of patients with selected tumor types experience durable clinical benefit. A need exists for more efficacious immuno-oncology approaches that are well tolerated.
- T cell regulation involves the integration of multiple signaling pathways: signaling via the TCR complex and through co-signaling receptors, both costimulatory and coinhibitory. One co-stimulatory pathway is activated by CD80, a transmembrane protein that is expressed on professional antigen-presenting cells such as dendritic cells and activated macrophages.
- Following TCR recognition of cognate peptide-MHC, CD80 acts as a co-stimulatory ligand via interactions with its receptor, CD28, expressed on T cells. In addition to signaling via CD28, CD80 also interacts with the co-inhibitory molecule CTLA-4 and has been reported to interact with PD-L1. Together, the costimulatory and coinhibitory ligands ensure both tolerance to self-antigens and the ability to mount an appropriate immune response to non-self antigens.
- FPT155, a soluble CD80 fusion protein, is being developed for the treatment of solid tumors. FPT155 acts through CD28 to co-stimulate T cell responses only in the presence of antigenic TCR signaling. In addition to acting as a CD28 agonist, FPT155 binds to CTLA-4, preventing it from competing for endogenous CD80 and thereby acting as a checkpoint inhibitor. FPT155 thus enhances CD28 signaling to promote T cell activation both directly, in the context of Fc-mediated co-presentation with TCR stimulus, and indirectly, in the context of endogenous CD80 expression.
- FPT155-001 is an ongoing phase 1a/1b study of FPT155 in patients with advanced solid tumors.

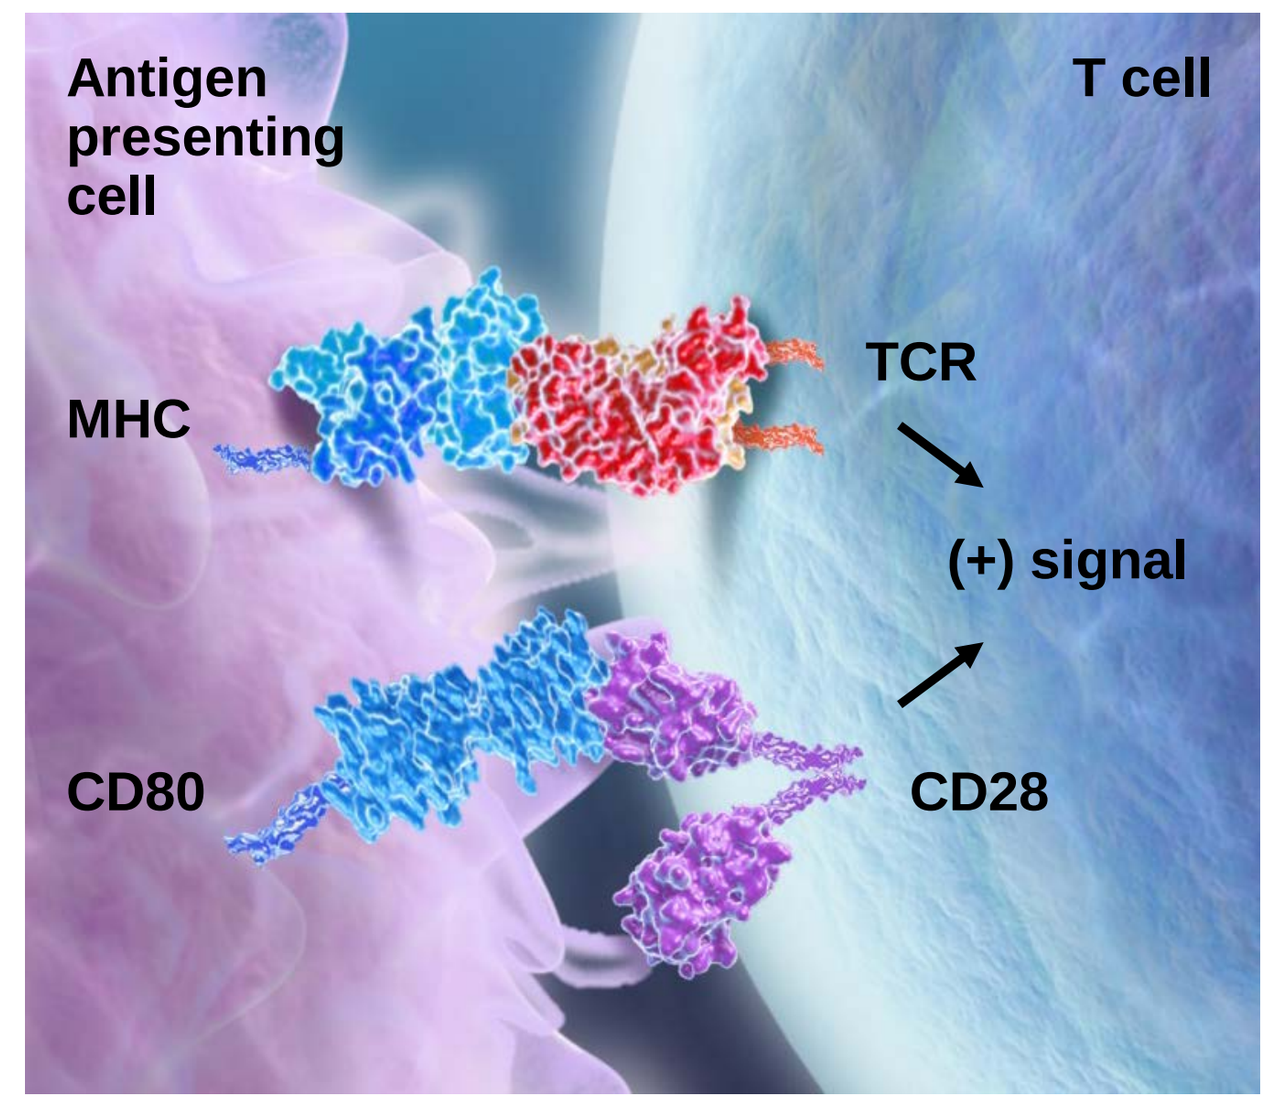
Antigen presenting cell
T cell
MHC
TCR
(+) signal
CD80
CD28

FPT155

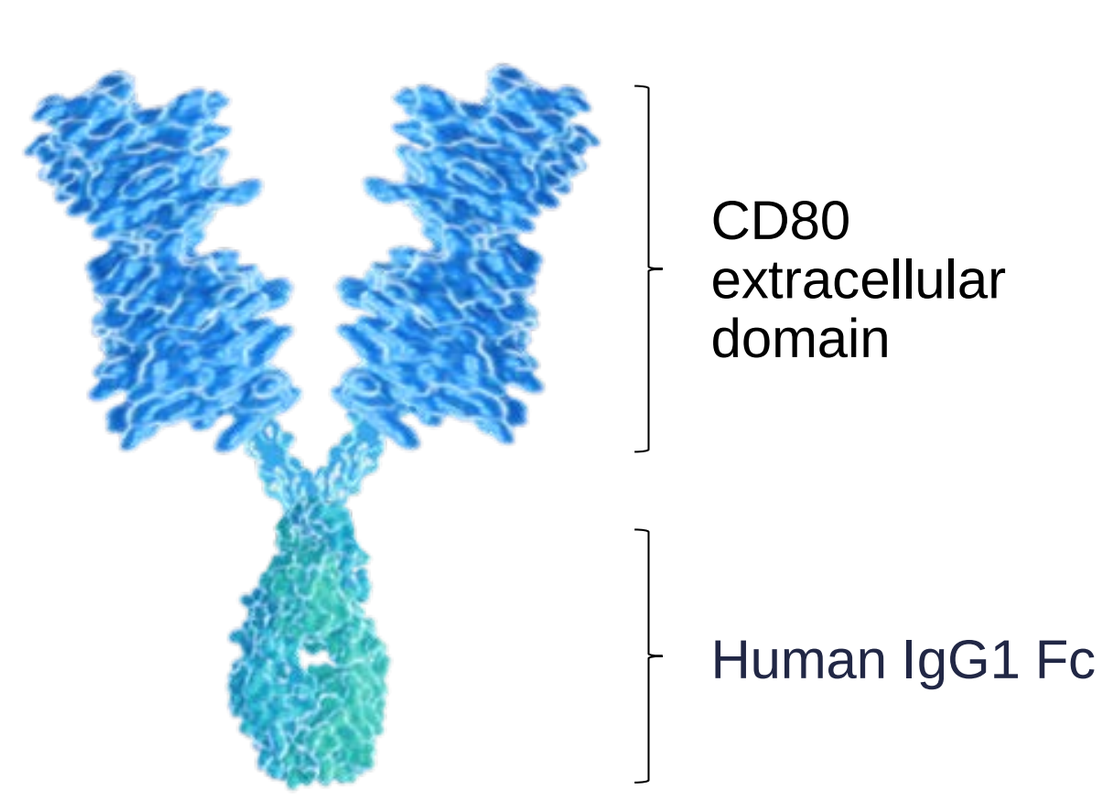
CD80 extracellular domain
Human IgG1 Fc

FPT155 Costimulates T Cells but Is Not a Superagonist

The Stebbings assay format predicts the cytokine release syndrome (CRS) induced in patients by the anti-CD28 superagonist antibody TGN1412. In this format, TGN1412 alone induces spontaneous cytokine release.

By contrast, FPT155 alone does not induce the spontaneous release of CRS-associated cytokines.

Robust cytokine production is observed when anti-CD3 stimulus is provided in combination with FPT155, demonstrating that FPT155 must be co-presented with TCR stimulus to activate T cells.

PBMC Spontaneous Cytokine Release *in vitro*

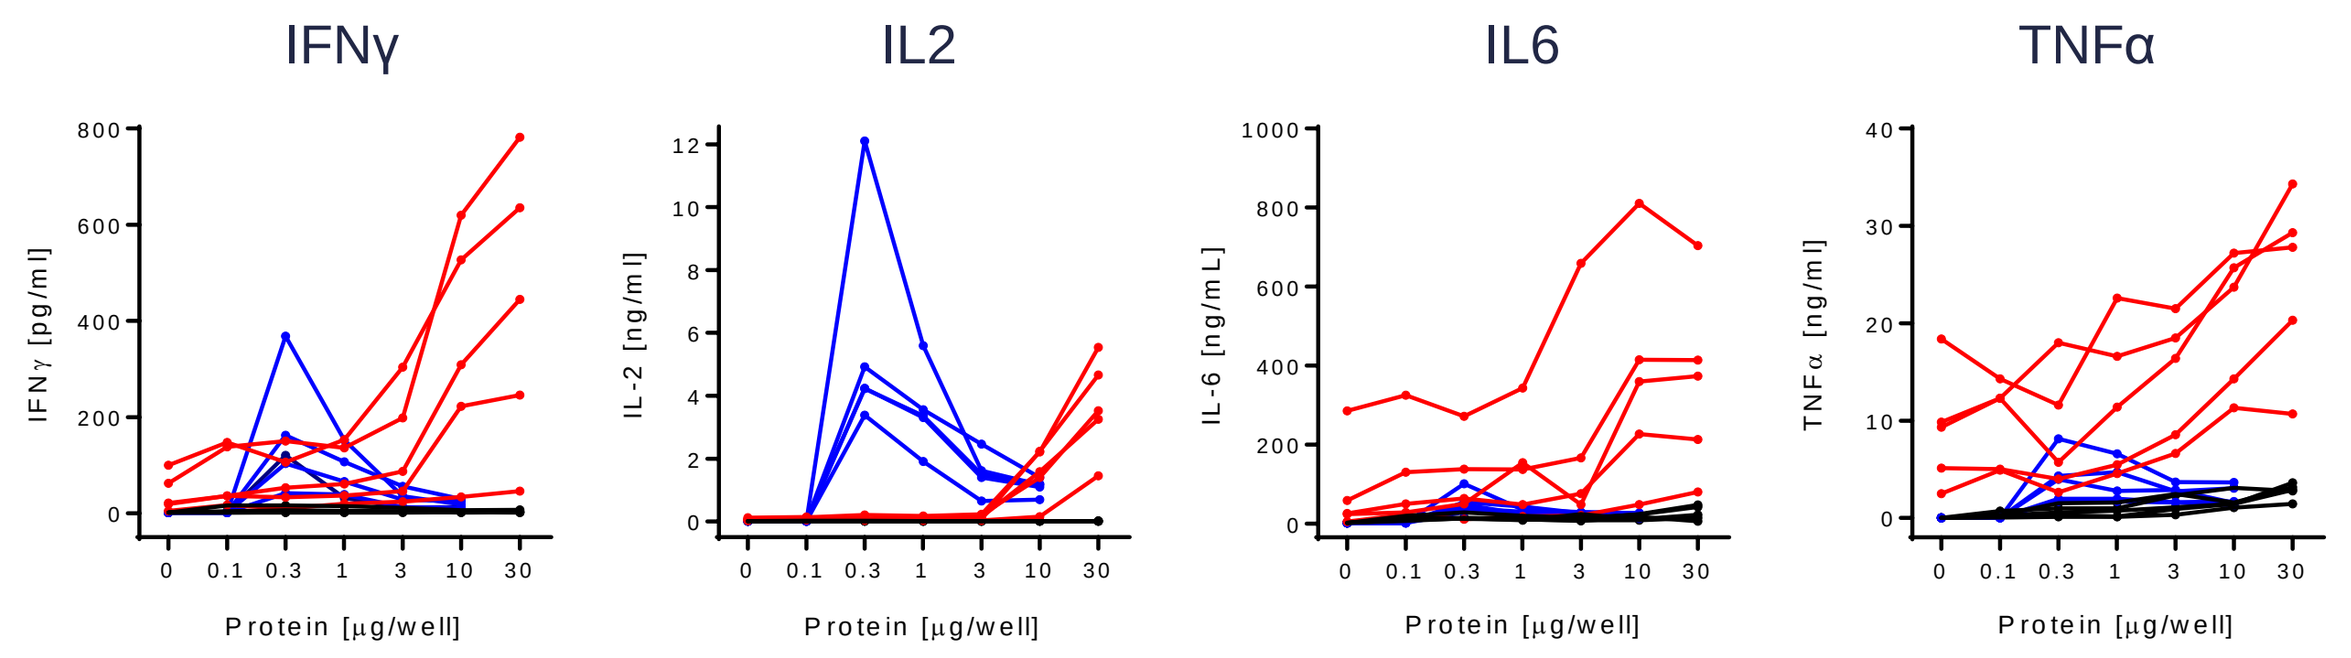
IFNγ
IL2
IL6
TNFα
IFNγ [pg/ml]
IL-2 [ng/ml]
IL-6 [ng/mL]
TNFα [ng/ml]
Protein [µg/well]
FPT155 alone
FPT155 + anti-CD3
TGN1412 alone

Each line represents an individual healthy donor (n = 5)

A Murine Surrogate, mFPT155, Induces Tumor Regressions at Low Doses

mFPT155 exerts dose-dependent inhibition of CT26 tumor growth following one administration at doses ≥0.1 mg/kg starting at an average tumor volume of 80-100 mm^3.

CT26 Tumor Growth

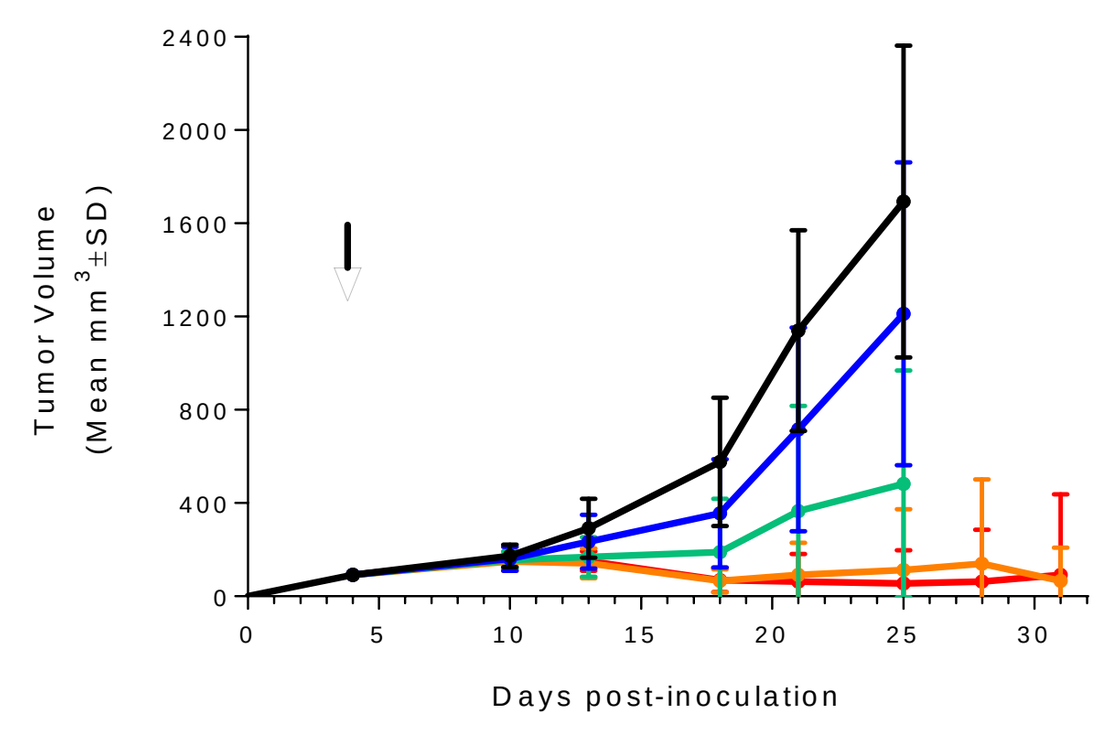
Tumor Volume (Mean mm³±SD)
Days post-inoculation

Day 21 Tumor Volume

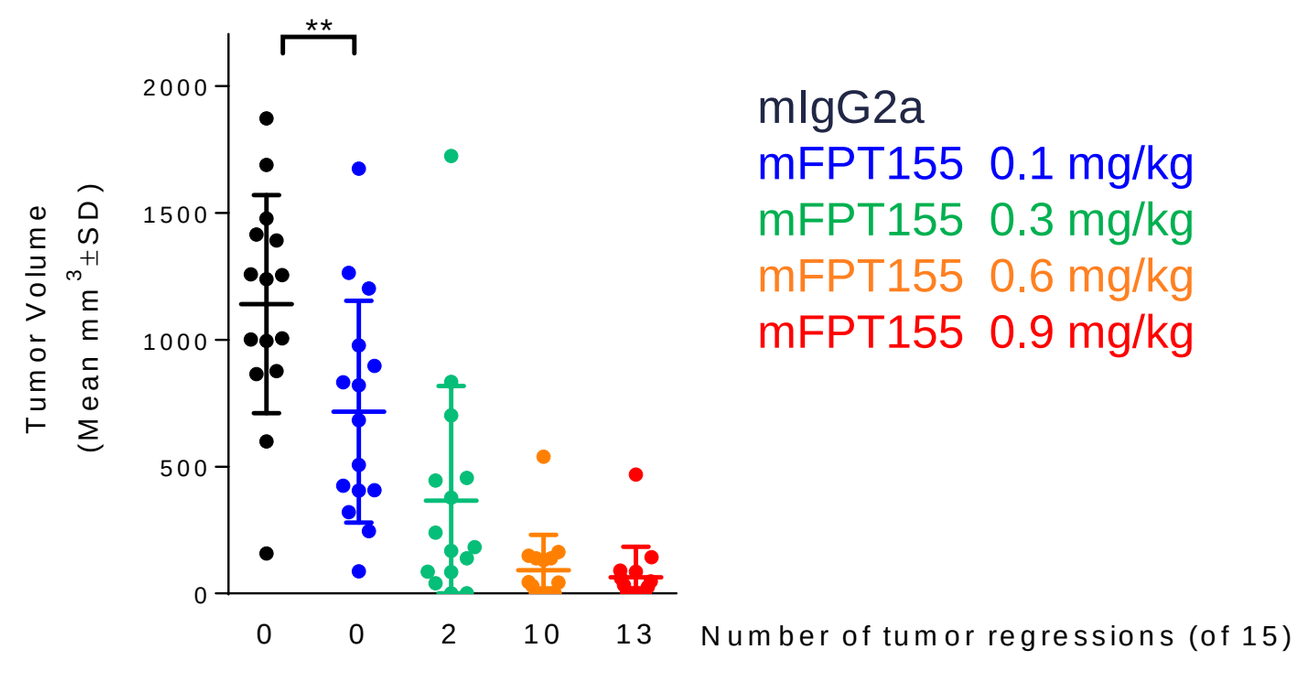
Tumor Volume (Mean mm³±SD)
**
0 0 2 10 13 Number of tumor regressions (of 15)
mIgG2a
mFPT155 0.1 mg/kg
mFPT155 0.3 mg/kg
mFPT155 0.6 mg/kg
mFPT155 0.9 mg/kg

Murine FPT155 (mFPT155) also demonstrates monotherapy anti-tumor activity in tumor models generally known to be refractory to anti-PD(L)1 and anti-CTLA4 agents: 4T1, and B16-F10

Mice re-challenged with CT26 tumors 8 weeks after clearance of initial primary tumor are fully protected from tumor growth.

No mFPT155 was administered during the second challenge.

CT26 Rechallenge Tumor Growth

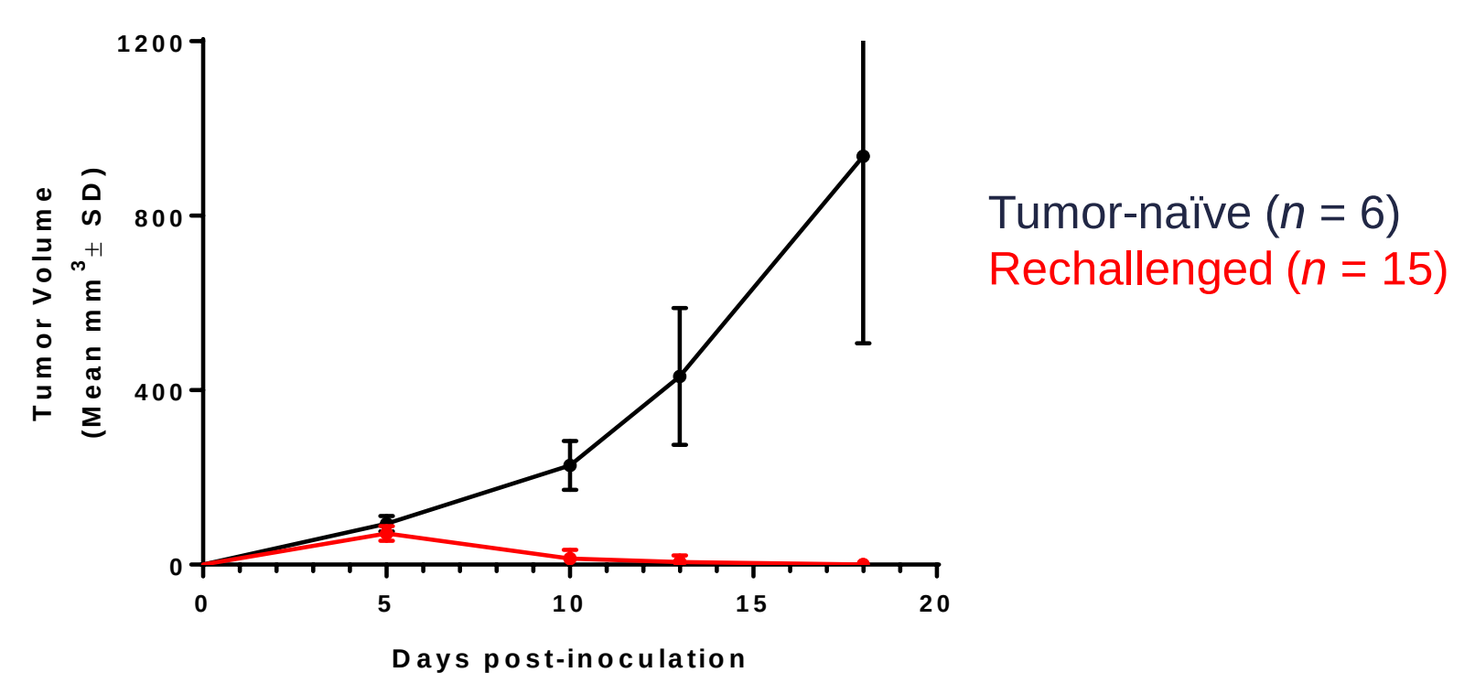
Tumor Volume (Mean mm³± SD)
Days post-inoculation
Tumor-naïve (n = 6)
Rechallenged (n = 15)

mFPT155 Promotes a Favorable Tumor Immune Contexture

Robust effector T cell infiltration is observed in CT26 tumors after two doses of mFPT155 at 0.3 mg/kg.

T Cell Recruitment to Tumors

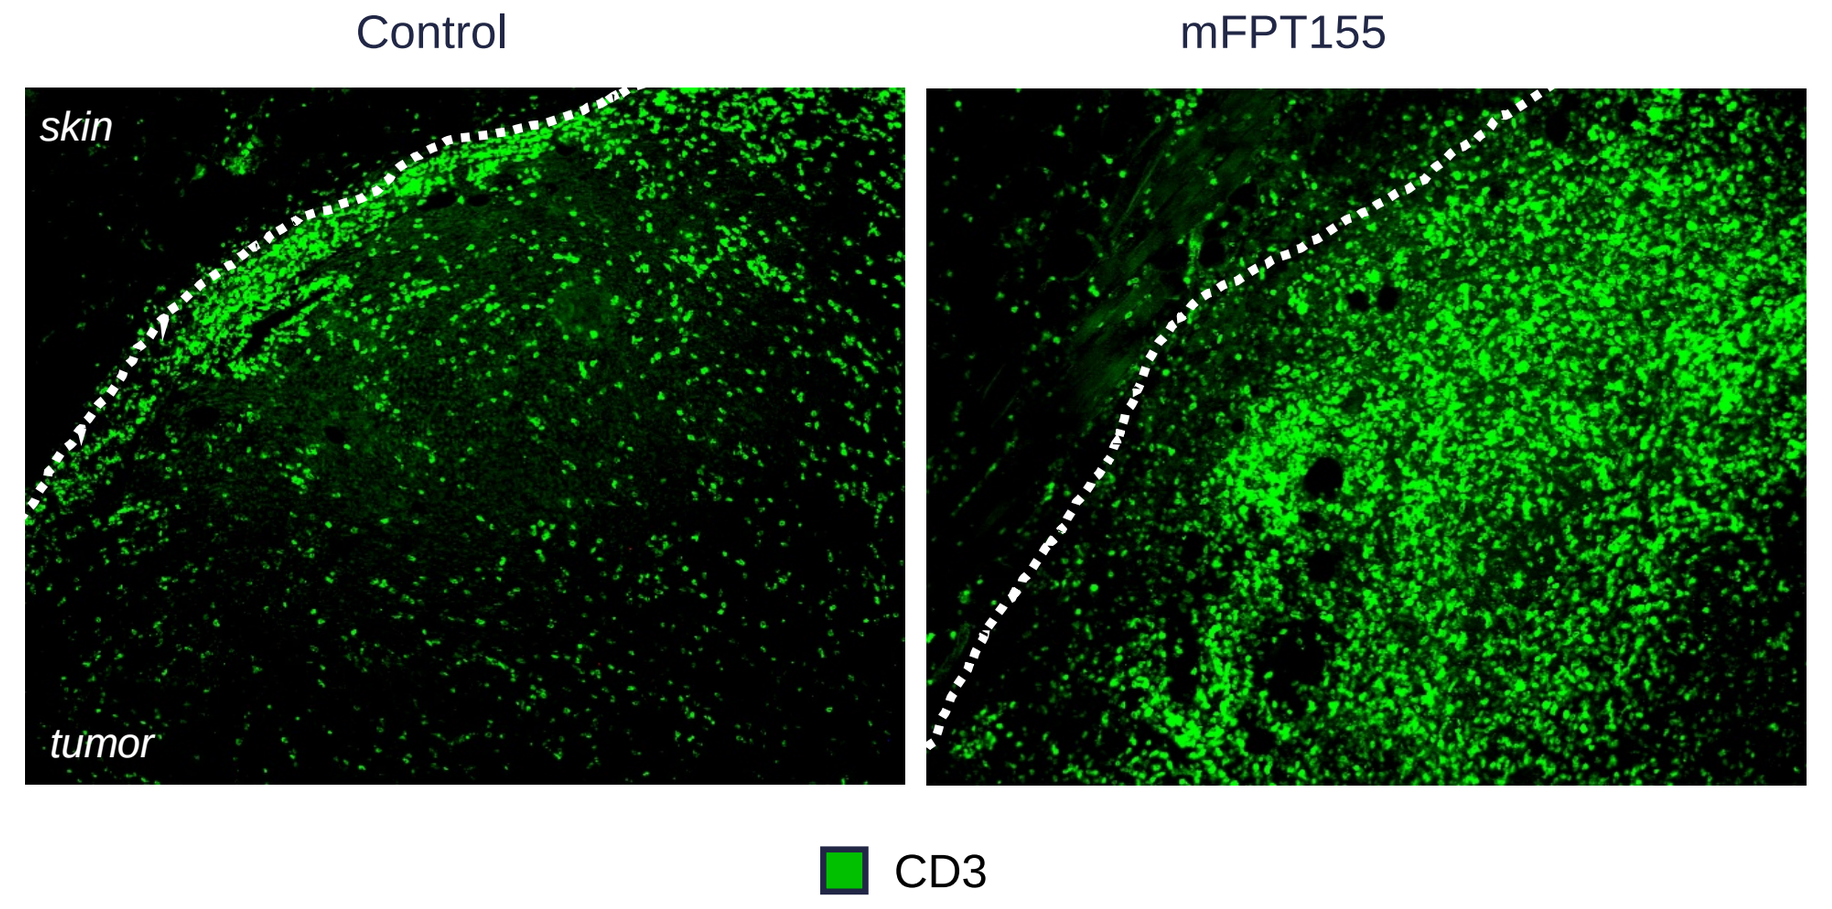
Control
mFPT155
skin
tumor
CD3

Ratio Between Effector T Cells and T_{reg}

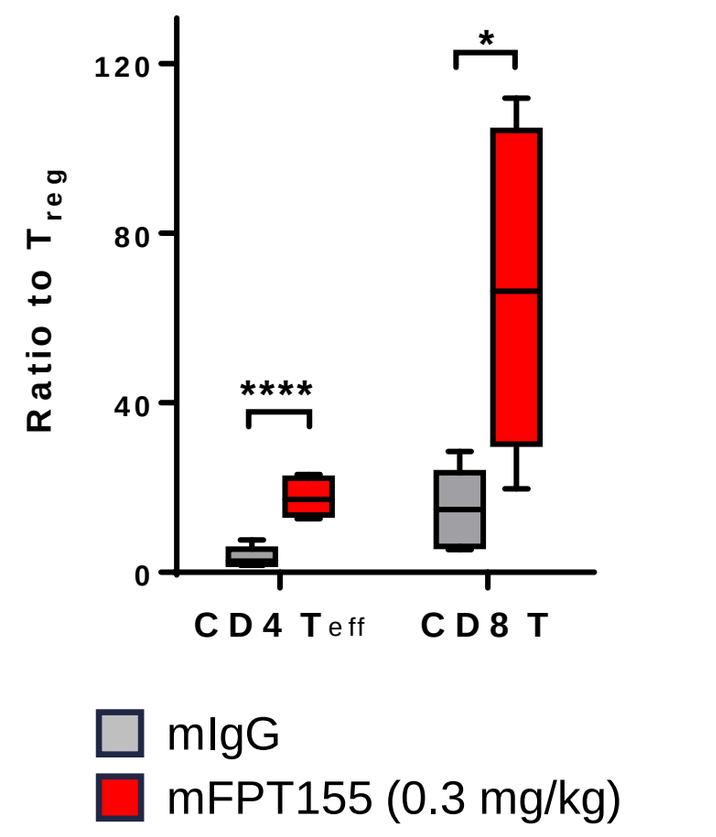
Ratio to Treg

*
CD4 Teff
CD8 T
mIgG
mFPT155 (0.3 mg/kg)

mFPT155 Has Synergistic Combination Activity with anti-PD1

CT26 Tumor Growth

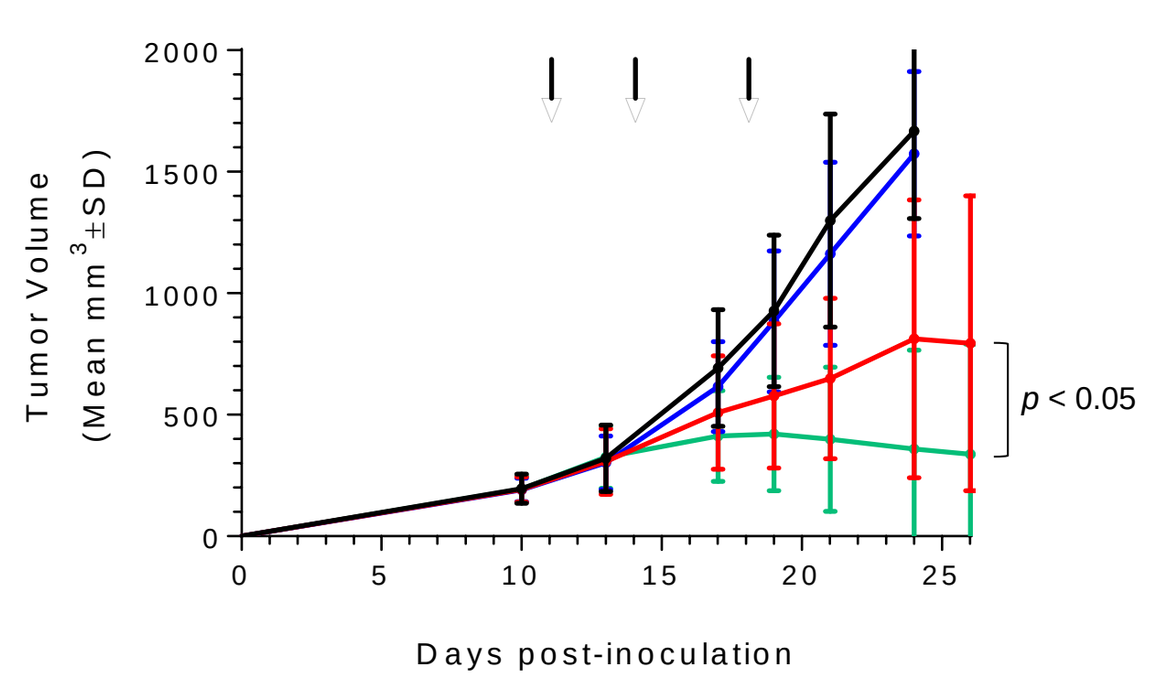
Tumor Volume (Mean mm³±SD)
p < 0.05
Days post-inoculation

Survival

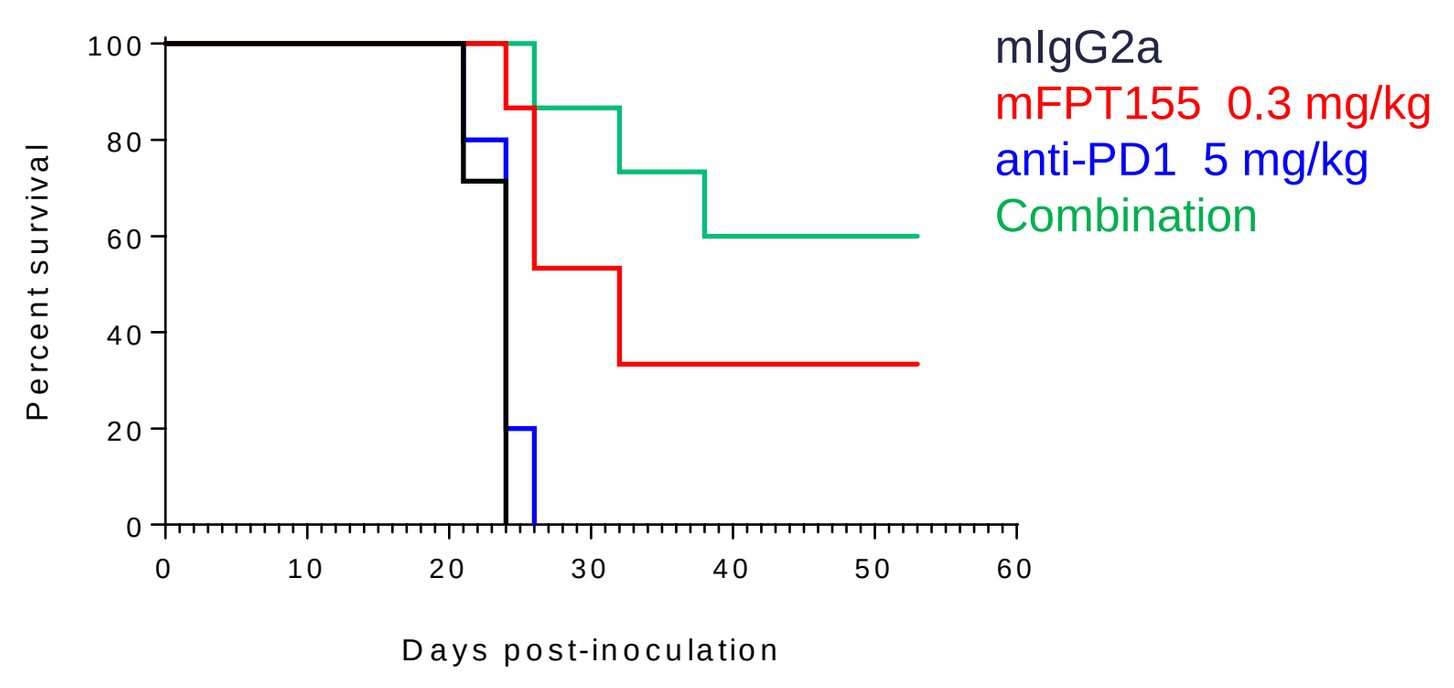
Percent survival
Days post-inoculation
mIgG2a
mFPT155 0.3 mg/kg
anti-PD1 5 mg/kg
Combination

Combination of FPT155 with anti-PD1 agent synergistically enhances tumor growth control. The combination regimen also increases the number of complete tumor rejections to 9 of 15, compared to 5 of 15 with mFPT155 alone.

Study Design

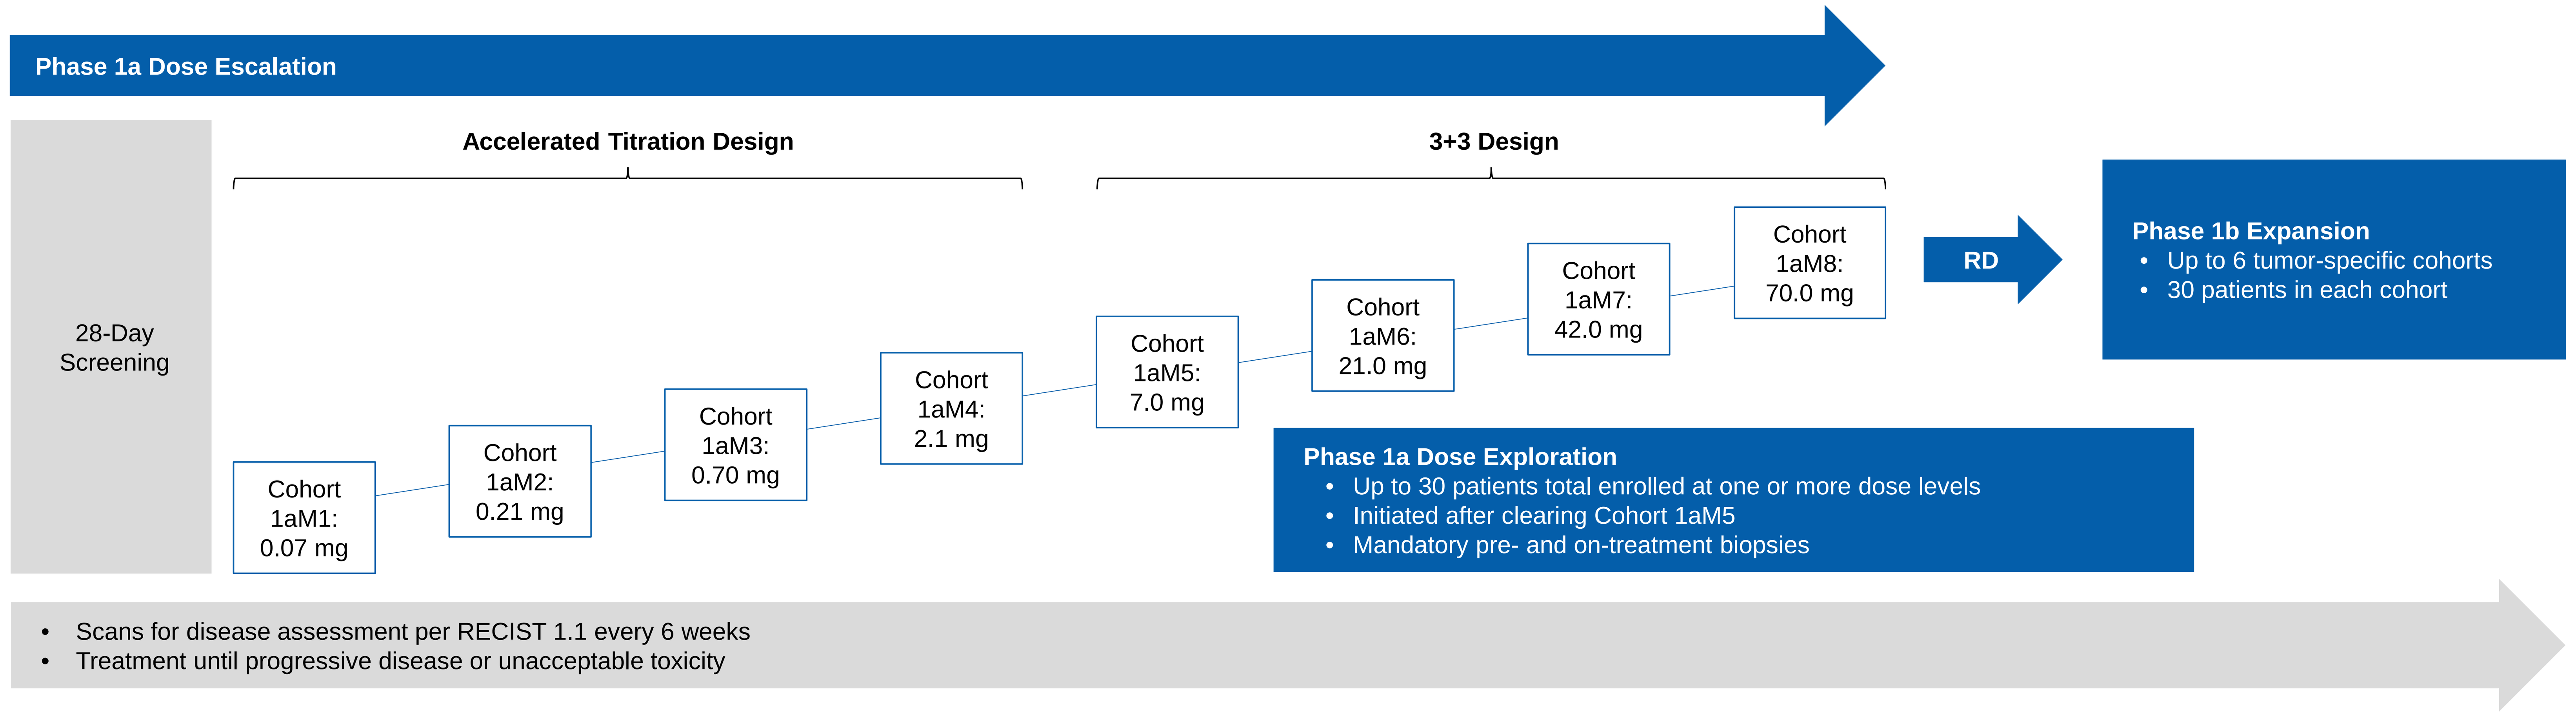
Phase 1a Dose Escalation
28-Day Screening
Accelerated Titration Design
3+3 Design
Cohort 1aM1: 0.07 mg
Cohort 1aM2: 0.21 mg
Cohort 1aM3: 0.70 mg
Cohort 1aM4: 2.1 mg
Cohort 1aM5: 7.0 mg
Cohort 1aM6: 21.0 mg
Cohort 1aM7: 42.0 mg
Cohort 1aM8: 70.0 mg
RD
Phase 1b Expansion
Up to 6 tumor-specific cohorts
30 patients in each cohort
Phase 1a Dose Exploration
Up to 30 patients total enrolled at one or more dose levels
Initiated after clearing Cohort 1aM5
Mandatory pre- and on-treatment biopsies
Scans for disease assessment per RECIST 1.1 every 6 weeks
Treatment until progressive disease or unacceptable toxicity

Study Objectives

Primary objectives

- To assess the safety and tolerability of FPT155 as monotherapy in patients with advanced solid tumors
- To determine the RD of FPT155 as monotherapy

Secondary objectives

- To characterize the PK profile of FPT155 as monotherapy in patients with advanced solid tumors
- To characterize the immunogenicity of FPT155 as monotherapy in patients with advanced solid tumors

Starting Dose Selection

- Minimum anticipated biologic effect level (MABEL) approach used to select safe starting dose
- FPT155 has approximately 1000x higher affinity for CTLA-4 than CD28
 - Starting dose of 0.07 mg (~0.001 mg/ kg) leads to ~40% receptor occupancy (RO) for CTLA-4 but < 1% RO for CD28
- Planned dose escalation will proceed at half-log intervals when CD28 engagement is low, with more conservative escalation (~2 fold) at higher CD28 RO

Determination of Dose-Limiting Toxicities

Any of the following deemed by the investigator as related to FPT155

- Absolute neutrophil count (ANC) < 1.0 x 10^9/L for > 5 days duration or Grade 3 febrile neutropenia (e.g., ANC < 1.0 x 10^9/L with a single temperature of > 38.3°C or fever > 38°C for more than 1 hour)
- Platelets < 25 x 10^9/L or platelets < 50 x 10^9/L with clinically significant hemorrhage
- Aspartate aminotransferase (AST) or alanine aminotransferase (ALT) > 3x upper limit of normal (ULN) with concomitant total bilirubin > 2x ULN not related to liver involvement with cancer
- Grade 3-4 non-hematological toxicity except:
 - Grade 3 fatigue
 - Grade 3 nausea and Grade 3-4 vomiting and diarrhea lasting < 72 hours in patients who have not received optimal anti-emetic and/or anti-diarrheal therapy
 - Grade 3 endocrinopathy that is adequately treated by hormone replacement
 - Laboratory value that may be corrected through replacement within 48 hours
- Grade 2 neurological toxicity except headache and peripheral neuropathy in patients with Grade 1-2 peripheral neuropathy at entry

Enrollment Criteria

Inclusion criteria

- Histologically confirmed solid tumors (except primary CNS tumors)
- Disease that is unresectable, locally advanced, or metastatic and has progressed following all standard treatments or is not appropriate for standard treatments
- All patients must have at least one measurable lesion at baseline according to RECIST v1.1. Tumor sites situated in a previously irradiated area, or in an area subjected to other loco-reginal therapy, are not considered measurable unless there has been demonstrated progression in the lesion.
- Availability of archival tumor tissue and consent to provide archival tumor for retrospective biomarker analysis, or consent to undergo a fresh tumor biopsy during screening
- For patients participating in Phase 1a dose exploration cohorts: consent to undergo a mandatory fresh tumor biopsy during screening and on treatment
- Adequate hematologic parameters and renal and liver function as defined in the protocol

Key exclusion criteria

- Treatment with any anti-cancer therapy or participation in another investigational drug or biologics trial within 28 days or ≤ 5 half-lives (whichever is shorter) prior to the first dose of study treatment administration or while on this study
- Prior treatment with a CTLA-4 antagonist, including ipilimumab or tremelimumab
- Patients who have received prior immune-modulating therapies (including regimens containing an immune agonist or a PD-L1 or PD-1 antagonist are NOT permitted to enroll unless all of the following apply:
 - Must not have experienced a drug-related toxicity that led to permanent discontinuation of prior immunotherapy
 - Treatment was last administered 5 half-lives or 90 days (whichever is shorter) prior to first dose of study treatment

Summary

FPT155 is a first-in-class CD80-Fc fusion protein engineered to activate T cells through multiple pathways

- The murine surrogate (mFPT155) has potent anti-tumor activity in multiple murine tumor models
- FPT155 does not have TCR-independent superagonist activity and does not elicit a cytokine release response *in vitro*

FPT155-001, a Phase 1a/1b study of FPT155 in patients with advanced solid tumors, is ongoing

- Open for enrollment in Australia with planned expansion into South Korea
- Planned expansion in the United States for Phase 1b

© 2019 Five Prime Therapeutics, Inc. All Rights Reserved