A High Content Screen to Identify Novel Factors That Restore Phagocytosis in COPD Alveolar Macrophages

Yiming Zhu[1]*, Jennifa Gosling[1]* Mehrdad Arjomandi[2], Joyce Kwan[1], Andrea Tham[2], Rachel Stiner[2], Karen Simpson[3] , Brian Wong[1], Simon Hall[3], Barbara Maschera[3], Edith M Hessel[3]
[1]Five Prime Therapeutics, Inc., Two Corporate Drive, South San Francisco, CA, USA
[2]Department of Medicine, University of California, San Francisco, CA, USA
[3]GlaxoSmithKline Medicines Research Centre, Gunnels Wood Road, Stevenage, Hertfordshire, UK
*These authors contributed equally to the research

Abstract

Rationale: COPD is a chronic progressive airway disease characterized by airway inflammation, bacterial colonization and irreversible air flow obstruction. Alveolar macrophages (AMs) from COPD patients have been reported to be defective in bacterial phagocytosis, which can contribute to increased bacterial colonization and increased severity and frequency of COPD exacerbations. We sought to develop an image-based, high-content screen to identify novel potential therapeutic targets that may enhance AM phagocytosis in COPD patients.
Methods: Healthy volunteers (never-smokers), smokers with normal spirometry (>20 pack-years of smoking), and patients with COPD (current and ex-smokers) with a history of exacerbations underwent bronchoscopy to obtain alveolar macrophages. The cells were analyzed by flow cytometry for a selected panel of surface markers and cultured for phagocytosis assessment. To screen for factors that could regulate phagocytosis, AMs were cultured in 384-well plates and each well was treated with supernatants containing one protein from FivePrime's proprietary library of extracellular proteins prior to challenge with bacteria. AMs were labelled with cytoplasmic and nuclear dyes and challenged with heat-killed bacteria conjugated with pHrodo, a pH-sensitive fluorescent dye that is activated in lysosomes. Cells were then fixed and imaged using an IN Cell Analyzer 2000 followed by multi-parameter analysis utilizing the IN Cell Developer software. Data analysis and protein activity determination was done using z-score-like statistics based on median, strictly standardized mean difference (SSMD) and reproducibility in samples from multiple COPD donors.
Results: Active smoking caused down regulation of cell surface marker expression while AMs from COPD patients exhibited a trend of altered cell surface phenotype compared to controls as assessed by flow cytometry. Phagocytosis of non-typeable Haemophilus influenza (NTHi), a disease-relevant bacteria, was defective in AMs from COPD ex-smokers vs. healthy never-smokers (N=6, p<0.05, student t-test). A protein enhancer of phagocytosis, represented in the library, consistently showed activity in multiple COPD AM donors (N=6, p<0.05, student t-test) demonstrating proof-of-concept of this assay system. This enhancer as well as cytochalasin D were selected as quality control markers for the screen. All 5700 human extracellular proteins from the FivePrime proprietary library were screened and multiple proteins were identified that both enhanced and further inhibited phagocytosis by COPD AMs.
Conclusion: We successfully developed and completed an image-based, high-content screen using primary human COPD AMs and discovered novel extracellular proteins that modulate phagocytosis. Further evaluation of these proteins and elucidation of their biology may lead to potential therapies for patients with COPD.

Introduction

Chronic Obstructive Pulmonary Disease (COPD) is a long-lasting airway disease characterized by progressive development of irreversible airflow limitation and is usually associated with persistent airway inflammation, chronic bronchitis with fibrosis and/or airway emphysema, and exacerbations [1,6]. Although there are genetic and epigenetic factors that can predispose an individual to COPD, the primary risk factor is long-term exposure to tobacco smoke, which causes approximately 80% of cases of COPD[1,5,6]. As part of the first line of host defense in the respiratory tract, alveolar macrophages (AMs) play an important role in immune surveillance, clearance of foreign pathogens and damaged host cells as well as mounting immune responses in the airway[1,2,5]. Recent studies have indicated that COPD AMs are defective in the phagocytosis of common bacterial pathogens (e.g., NTHi, *Moraxella catarrhalis* and *Streptococcus pneumoniae*) found in these patients, which may contribute to bacterial colonization in the lower airway and increased bacterial infection leading to exacerbations in COPD[3,4,6,7,8]. Therefore, in this study we focused on identifying novel biological factors in the FivePrime proprietary library of extracellular proteins that may rescue the defective phagocytosis in COPD AM using a high-content screening approach.

Methods

Alveolar macrophage preparation: Based on their smoking status and disease status, healthy volunteers and COPD patients with a history of exacerbation were recruited and separated into four donor groups: healthy never-smokers, smokers with normal spirometry (with at least >20 pack-years smoking history), COPD ex-smokers, and COPD current smokers. After bronchoscopy, AMs were isolated from the bronchoalveolar lavage fluid using multiple washes, and filtration to remove mucus. This study was approved by the IRB of the University of California, San Francisco.

Bacteria labeling and preparation: NTHi (ATCC 53600) was cultured in Brain Heart Infusion broth to obtain a 0.6 OD_{600}. The culture media was removed, bacteria were re-suspended in PBS, and heat killed for 2 hours at 60°C. After heat-killing, the bacteria were pelleted by centrifugation and lyophilized overnight. The dry pellet was weighed, and re-suspended to 20mg/ml in sodium bicarbonate buffer. Labeling of the bacteria was performed using pHrodo Red SE labeling kit (P36600) from Invitrogen per the manufacturer's instructions.

Surface marker assessment using FACS: Alveolar macrophages (approximately 1-5x10^5 cells) in 100ul FACS buffer (PBS with 0.1% BSA) were stained with 5ul of APC-conjugated antibodies specific for CD71 (BD#550854**)**, CD14 (eBio#17-0149), CD16 (BD#561304), CD163 (Biolegend#333610), CD1a (BD#559775), CD1b (eBio#17-0018-42) and the corresponding isotype control (BC#550854) for 30min in the dark. Cells were then washed once with FACS buffer, and re-suspended in 100ul of FACS buffer prior to analysis using a LSRII flow cytometer.

Phagocytosis assay and high throughput screening: Within 4 hours of bronchoscopy, AMs were plated at a density of 3000 cells/well in a 384-well optically clear plate with RPMI and 10% heat-inactivated FBS. Cells were cultured overnight at 37°C, 5% CO_2. After media change, each well was treated with a supernatant containing a protein from FivePrime's proprietary library of extracellular proteins transiently expressed in 293 cells. After 18 hours of treatment, the cells were washed and stained with CellTracker Green and Hoechst per the manufacturer's instruction. pHrodo-labeled heat-killed NTHi was added to the designated wells and incubated for 2hr at 37°C . Cells were fixed prior to imaging using the INCell Analyzer 2000. Images, containing ~75 cells/field, were analyzed with a multi-parameter protocol using INCell developer software. Illustration of the analysis algorithm and examples of segmentation are shown below. Data analysis and protein activity was determined using z-score-like statistics based on median, strictly standardized mean difference (SSMD) and reproducibility in samples from multiple COPD donors.

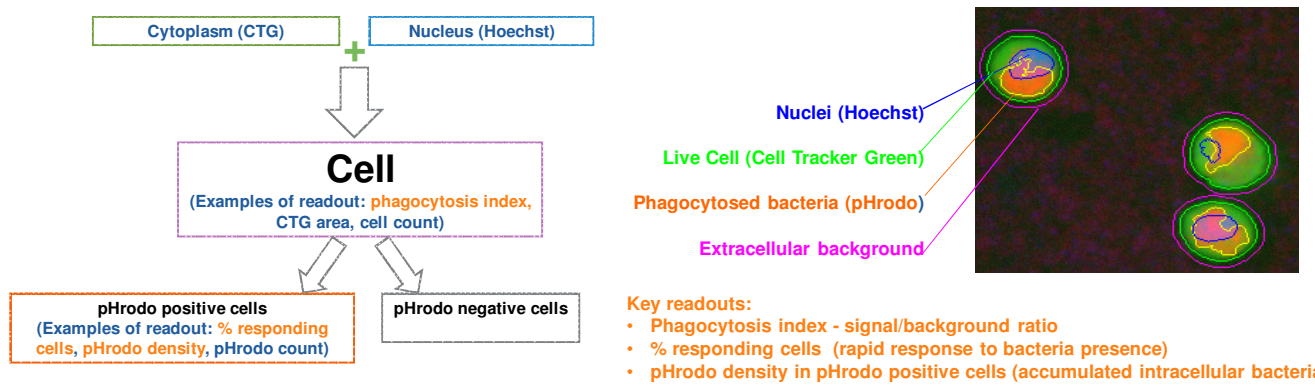
Cytoplasm (CTG)
Nucleus (Hoechst)
Cell
(Examples of readout: phagocytosis index, CTG area, cell count)
pHrodo positive cells
(Examples of readout: % responding cells, pHrodo density, pHrodo count)
pHrodo negative cells
Nuclei (Hoechst)
Live Cell (Cell Tracker Green)
Phagocytosed bacteria (pHrodo)
Extracellular background

Key readouts:
- Phagocytosis index - signal/background ratio
- % responding cells (rapid response to bacteria presence)
- pHrodo density in pHrodo positive cells (accumulated intracellular bacteria)

Results

Donor Group	Donor Numbers	Male/Female	Age Yrs (mean ± SD)	FEV1 (% predicted) (mean ± SD)	Gold Stage 1/2/3/4
Healthy never-smoker	10	8/2	61 ± 6.4	101 ± 10.8	N/A
Smoker with normal spirometry	10	8/2	54 ± 4.8	79 ± 14.8	N/A
COPD ex-smoker	8	5/3	69 ± 4.0	52 ± 14.3	0/5/3/0
COPD smoker	14	12/2	64 ± 7.9	56 ± 19.1	1/10/0/3

Table 1: Demographic characteristics of participants

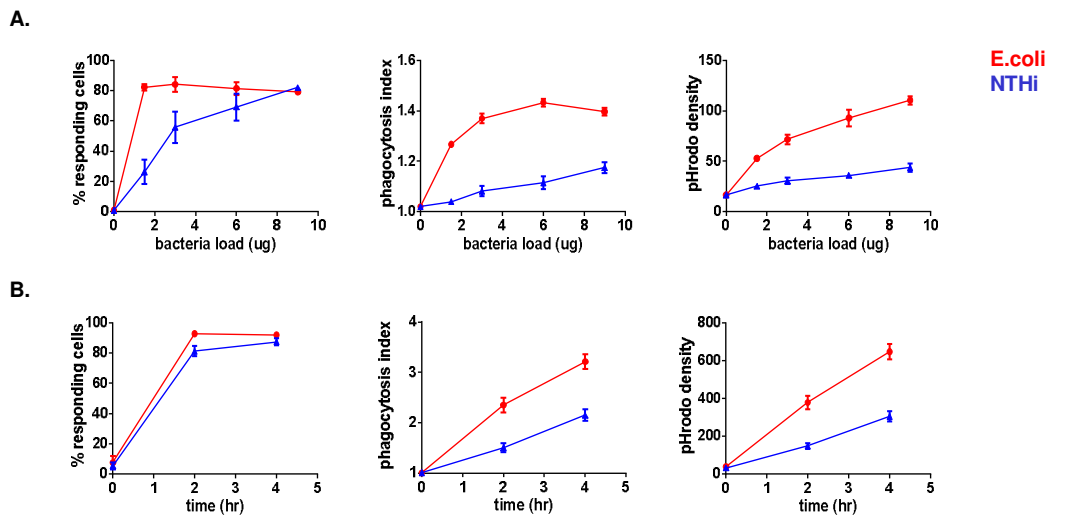
A.
B.
% responding cells
phagocytosis index
pHrodo density
bacteria load (ug)
time (hr)
E.coli
NTHi

Figure 1: Bacterial uptake in AMs occurred in a dose- and time-dependent manner Alveolar macrophages from a COPD smoker were challenged with in-house labeled pHrodo-NTHi or commercial pHrodo-E.coli for 3 hours. Increase in uptake was measured by an increase in signal in all three key readouts: % responding cells, phagocytosis index and pHrodo density (A). In addition, time-dependent bacterial uptake in COPD AMs was observed up to 4 hours post challenge (B). Signal to background [(RFU at pH4) / (RFU at pH7)] was comparable for the two labeled bacteria (data not shown). Similar effects where obtained with other bacteria such as M. catarrhalis and S. aureus (data not shown). Mean ± SD shown.

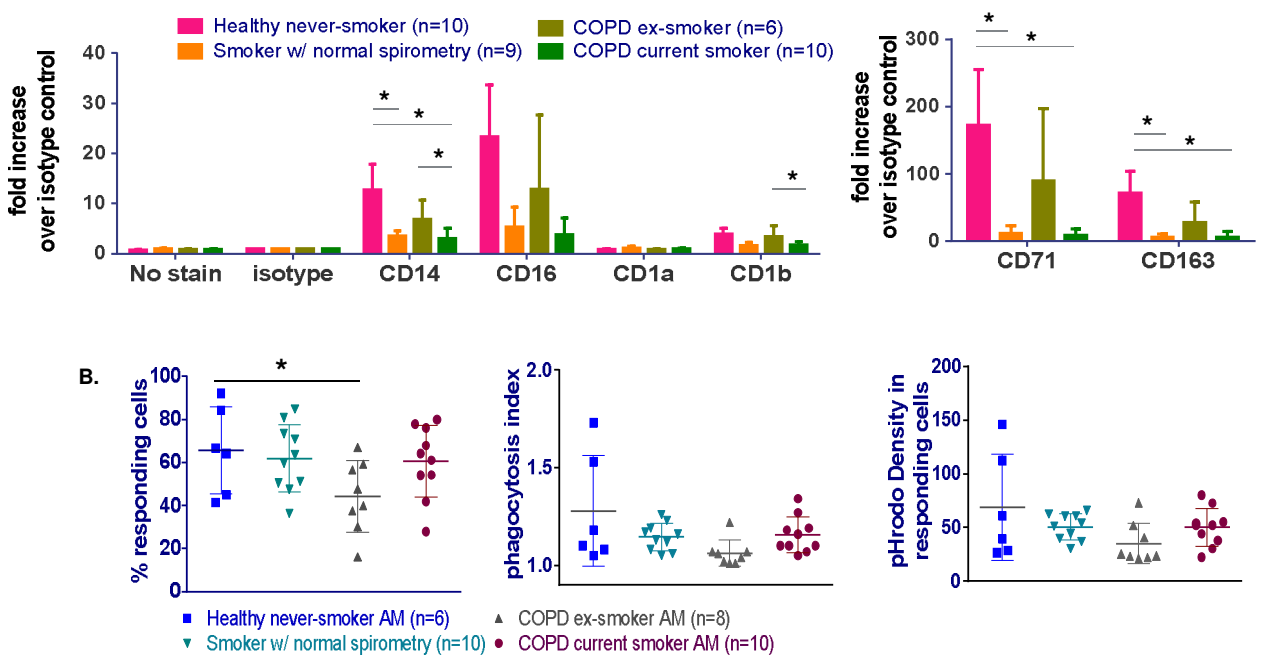
A.
Healthy never-smoker (n=10)
Smoker w/ normal spirometry (n=9)
COPD ex-smoker (n=6)
COPD current smoker (n=10)
fold increase over isotype control
No stain
isotype
CD14
CD16
CD1a
CD1b
CD71
CD163
B.
% responding cells
phagocytosis index
pHrodo Density in responding cells
Healthy never-smoker AM (n=6)
Smoker w/ normal spirometry (n=10)
COPD ex-smoker AM (n=8)
COPD current smoker AM (n=10)

Figure 2: Differential expression of surface markers and uptake of pHrodo-NTHi was observed in different donor groups. Significant decreases in CD71, CD14 and CD163 were observed in smokers with normal spirometry and COPD smokers compared to the healthy never-smokers (*p<0.05, student t test). In COPD smokers, significant decreases in CD14 and CD1b were detected compared to those of COPD ex-smokers (*p<0.05, student t test). These results suggest an effect of active smoking on these markers in both smokers with normal spirometry and COPD smokers. (A). To understand the difference in bacteria uptake among these donor groups**,** AMs were challenged with 6ug of pHrodo-NTHi for 3 hours**.** Uptake of the bacteria was analyzed in all four donor groups and a significant decrease was observed in COPD ex-smokers vs. healthy never-smokers using the % responding cells readout (*p<0.05, student t test) (B). This is consistent with previous reports[3,4,7,8] that COPD AMs show defective phagocytosis compared to healthy AMs. Mean ± SD shown.

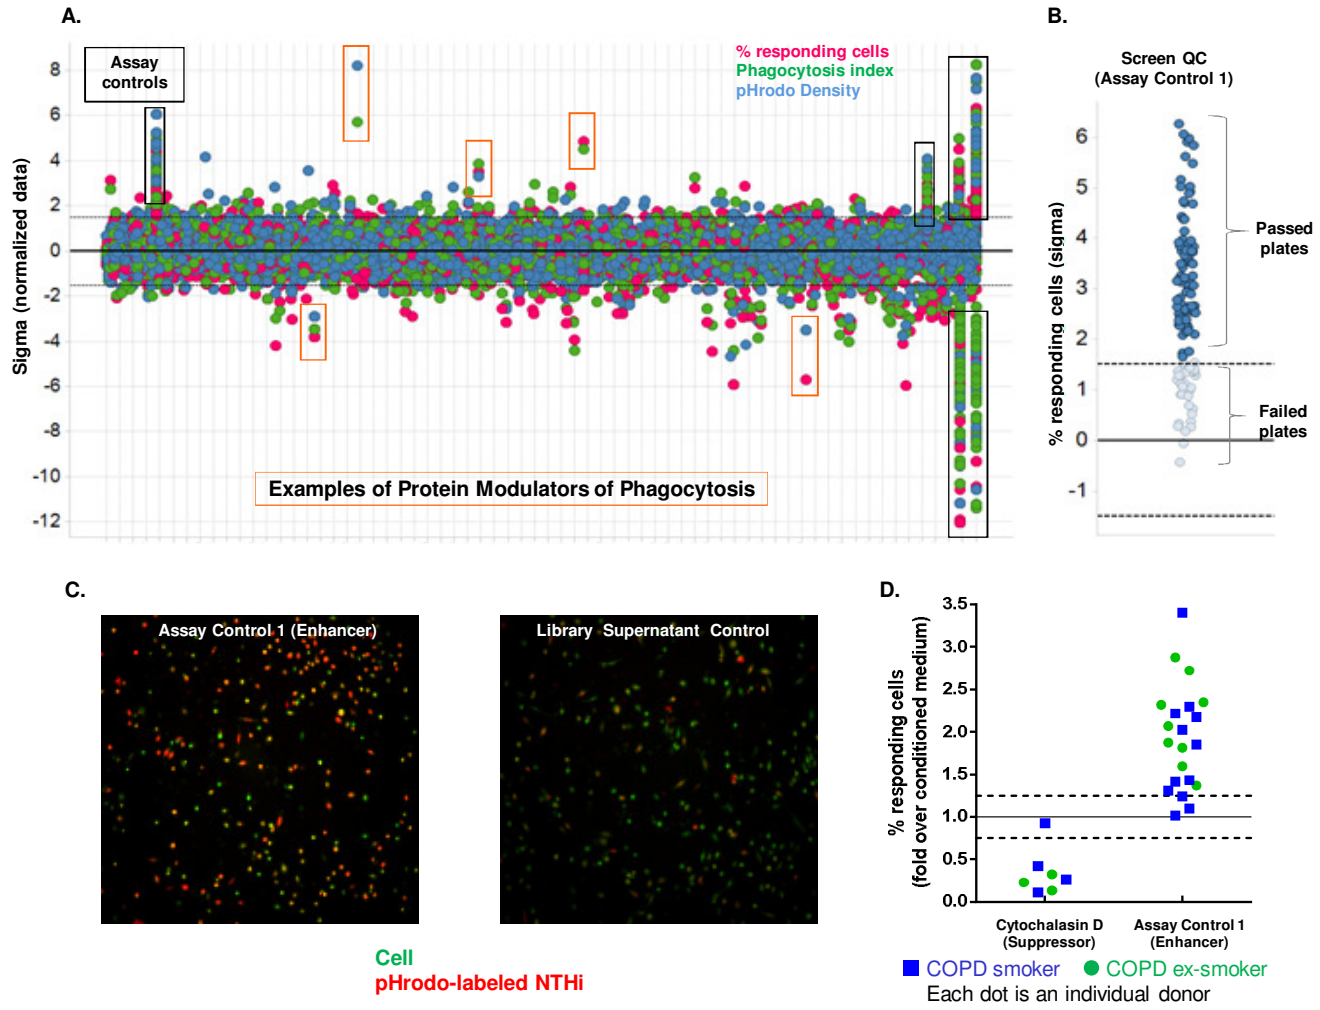
A.
Assay controls
% responding cells
Phagocytosis index
pHrodo Density
Sigma (normalized data)
Examples of Protein Modulators of Phagocytosis
B.
Screen QC
(Assay Control 1)
% responding cells (sigma)
Passed plates
Failed plates
C.
Assay Control 1 (Enhancer)
Library Supernatant Control
Cell
pHrodo-labeled NTHi
D.
% responding cells
(fold over conditioned medium)
Cytochalasin D
(Suppressor)
Assay Control 1
(Enhancer)
COPD smoker
COPD ex-smoker
Each dot is an individual donor

Figure 3: High content imaging screen identified potential protein modulators of AM phagocytosis. Screening data on over 5700 extracellular proteins were normalized to the plate median and represented as sigma (standard deviation from the plate median) for each of the three key readouts. A threshold for protein activity of 1.5 sigma (dotted line) was selected based on the effect size of the assay controls. Examples of proteins with activity (red boxes) demonstrate the dynamic range of the screen (A). Panel B shows a summary of the screen QC across all assay plates based on one of the assay controls, Assay Control 1 (each dot represents a single assay plate). Assay plates were invalidated and rescreened when Assay Control 1 failed to enhance uptake above 1.5 sigma (transparent dots). A failure rate of approximately 30% was observed across the entire screen (B). An image example of Assay Control 1 that enhanced phagocytosis and control supernatant (baseline) are shown. An increase in red fluorescence in the image of Assay Control 1 compared to that of control supernatant demonstrates an elevated uptake of bacteria (C). Proteins that modulated phagocytosis were retested in multiple COPD donors to assess the reproducibility of the activity and donor variability. Results of the multi-donor study of Cytochalasin D and Assay Control 1 are illustrated in Panel D. Activities of Cytochalasin D and Assay Control 1 were reproduced in multiple COPD donors (including current smokers and ex-smokers), validating the results from the primary screen (D).

Conclusions and Future Directions

We observed differential expression levels of selected surface markers on alveolar macrophages from COPD and healthy individuals, suggesting suppressive effects of active smoking on CD71 (differentiation marker), CD14 (monocytic marker) and CD163 (marker for M2a macrophages). Additionally, we demonstrated defective phagocytosis of bacteria as % of responding cells, but not other readouts, in AMs from COPD ex-smokers compared to AMs from healthy never-smokers using a pHrodo-labeled bacteria. We successfully developed and completed a high content screen of bacterial uptake in COPD alveolar macrophages using the FivePrime proprietary protein library. Numerous proteins were identified from our primary screen that showed consistent modulatory effects on phagocytosis in AMs from multiple COPD donors. These proteins are currently being evaluated in follow-up studies for potential therapeutic development for COPD.

References

1. **P.J. Barnes, Clin Chest Med 35 (2014)**
2. **T. Hussel and T. Bell, Nature Rev. Immunol. (2014)**
3. **C.S. Berenson *et.al.*, J Infectious Disease (2013)**
4. **A.E. Taylor *et. al.*, Eur Respir J (2010)**
5. **P.J. Barnes, Pharmacol. Rev. (2004)**
6. **A. Papi et al., Am J Respir Crit Care Med (2006)**
7. **S.D. Ramsey and F.D. Richard Hobbs, Proceeding of American Thoracic Society (2006)**
8. **L.E. Donnelly and P.J. Barnes, Chest (2012)**
9. **C.S. Berenson *et.al.*, J Infectious Disease (2006)**

Acknowledgements

We would like to thank our collaborators, Dr. Arjomandi and his lab, for recruitment, bronchoscopy and preparation of the alveolar macrophages as well as for scientific discussions. GlaxoSmithKline provided funding for this research.

© 2015 Five Prime Therapeutics, Inc. All Rights Reserved.